P0366 Burden of All-cause and Norovirus-associated Medically Attended Acute Gastroenteritis Among Adults with Underlying Medical Conditions in Germany, 2015–2024

Christine Kim, PhD, MSPH[1]; Worku B. Ewnetu, PhD[2]; Scott A. McDonald, PhD[2]; Wen-Hsing Wu, MS[1]; Chukwuemeka Onwuchekwa, MBBS, MSc[2]; Mikel Esnaola, MSc, PhD[2]; Jehidys Montiel, MSc, PhD[2]; Raeleesha Norris, MPhil[4]; Vukašin Višković, MA[4]; Brandon J. Patterson, PharmD, PhD[1]; Jennifer Radin, PhD, MPH[1]; Julia M. Baker, PhD, MPH[3]; Stefan Scholz, PhD, MPH[5]; Cornelia Voigt, PhD[5]; Shehzad M. Iqbal, PhD, MBA[1]; Katherine B. Carlson, PhD, MPH[1]

[1]Moderna, Inc., Cambridge, MA, United States; [2]P95 Clinical and Epidemiology Services, Leuven, Belgium; [3]Epidemiologic Research & Methods, LLC , United States; [4]InGef - Institute for Applied Health Research Berlin GmbH, Berlin, Germany; [5]Moderna Germany GmbH, Munich, Germany

*Presenting author.

BACKGROUND

- Norovirus is the leading cause of acute gastroenteritis (AGE) globally among adults, with substantial burden particularly among older adults and adults with underlying medical conditions.[1,2]
- In Germany, laboratory-confirmed norovirus infections are notifiable to the Robert Koch Institute, and norovirus diagnosis is directly linked to reimbursement of inpatient costs.[3]

OBJECTIVES

- This study aimed to update estimates of the adult burden of all-cause AGE and norovirus AGE across outpatient and inpatient settings by age group and presence of underlying medical conditions.

METHODS

Study design and data

- We conducted a retrospective cohort study using data from InGef, which includes de-identified medical claims from >10 million individuals in Germany, covered by ~50 statutory health insurance funds.
- All-case AGE and norovirus AGE encounters ≥14 days apart were identified via ICD-10-German Modification codes between July 1, 2015–June 30, 2024.
- Demographic characteristics and underlying medical conditions were assessed during the year prior to each season (July 1–June 30) of analysis.

Analysis

- Incidence of-AGE and norovirus AGE encounters was estimated using person-time methods; norovirus AGE was estimated using an indirect modeling approach, adapted from prior studies, which attributes cause-unspecified AGE episodes to norovirus to account for underdetection.[1]
- Incidence rates were estimated by age, healthcare setting, and select underlying medical conditions; 95% confidence intervals (CIs) were derived from bootstrapping. Rates were also estimated for individuals with specific underlying conditions (cardiovascular disease, chronic kidney disease, immunocompromising conditions, chronic gastrointestinal disease, diabetes, and blood disorders). Categories were not mutually exclusive; individuals could contribute to multiple condition groups.

RESULTS

Figure 1. Observed Mean Monthly Incidence Rate of All-cause AGE by Age Group, Stratified by Healthcare Setting

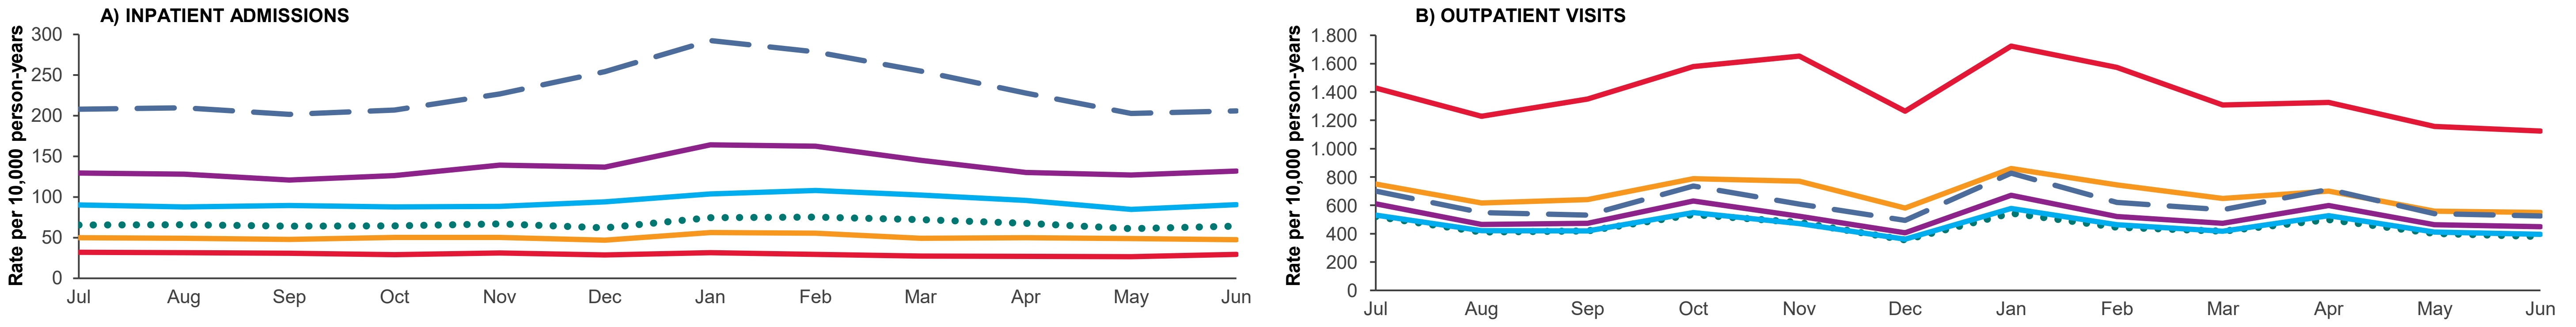
18-59
65-69
75-79
60-64
70-74
≥80
A) INPATIENT ADMISSIONS
Rate per 10,000 person-years
300
250
200
150
100
50
0
Jul
Aug
Sep
Oct
Nov
Dec
Jan
Feb
Mar
Apr
May
Jun
B) OUTPATIENT VISITS
1.800
1.600
1.400
1.200
1.000
800
600
400
200
0

Figure 2. Modeled Mean Monthly Incidence Rate of Norovirus AGE by Age Group, Stratified by Healthcare Setting

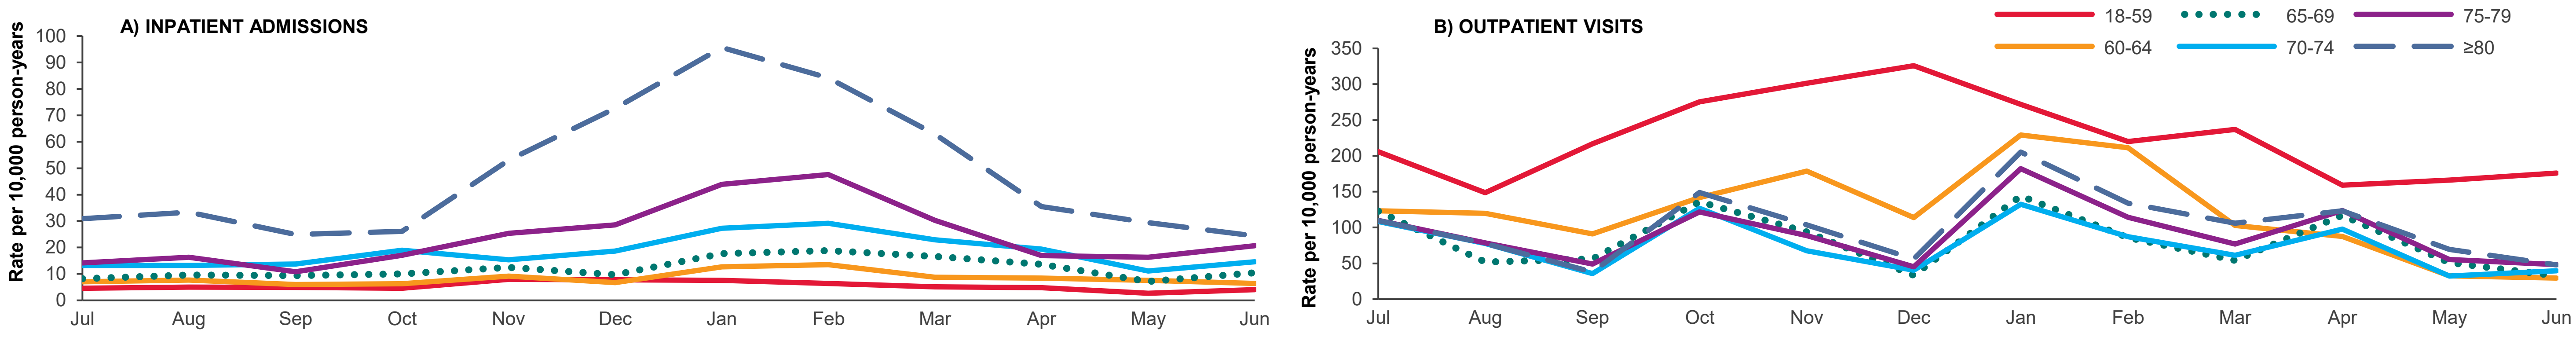
A) INPATIENT ADMISSIONS
Rate per 10,000 person-years
100
90
80
70
60
50
40
30
20
10
0
Jul
Aug
Sep
Oct
Nov
Dec
Jan
Feb
Mar
Apr
May
Jun
B) OUTPATIENT VISITS
18-59
65-69
75-79
60-64
70-74
≥80
350
300
250
200
150
100
50

Table 1. Demographic Characteristics and Underlying Medical Conditions among All-cause and Norovirus-associated AGE encounters in the InGef database by Healthcare Setting

	All-cause AGE		Norovirus AGE	
	Inpatient admissions n (%)	Outpatient visits n (%)	Inpatient admissions n (%)	Outpatient visits n (%)
Total number of patients	219,476	2,497,692	15,458	15,918
Total episodes	255,422 (100)	5,738,432 (100)	15,727 (100)	20,149 (100)
Age group				
18-59 years	105,178 (41.2)	4,948,629 (86.2)	5,482 (34.9)	16,905 (83.9)
60-64 years	20,265 (7.9)	276,328 (4.8)	944 (6.0)	1,017 (5.1)
65-69 years	20,735 (8.1)	139,750 (2.4)	1,036 (6.6)	510 (2.5)
70-74 years	21,783 (8.5)	107,754 (1.9)	1,244 (7.9)	351 (1.7)
75-79 years	27,028 (10.6)	103,696 (1.8)	1,802 (11.5)	396 (2.0)
≥80 years	60,433 (23.7)	162,275 (2.8)	5,219 (33.2)	970 (4.8)
Sex				
Male	114,201 (44.7)	2,978,136 (51.9)	7,255 (46.1)	9,869 (49.0)
Female	141,221 (55.3)	2,760,296 (48.1)	8,472 (53.9)	10,280 (51.0)
Number of underlying medical conditions				
0 (none)	29,487 (11.5)	1,681,228 (29.3)	1,703 (10.8)	5,909 (29.3)
1 condition only	40,940 (16.0)	1,735,517 (30.2)	2,442 (15.5)	5,924 (29.4)
≥2 conditions	184,995 (72.4)	2,321,687 (40.5)	11,582 (73.6)	8,316 (41.3)
Any underlying medical condition*				
Blood disorders	17,165 (6.7)	98,207 (1.7)	1,028 (6.5)	472 (2.3)
Cardiovascular disease	170,285 (66.7)	1,617,724 (28.2)	11,077 (70.4)	6,103 (30.3)
Chronic respiratory disease	107,152 (42.0)	2,089,213 (36.4)	6,588 (41.9)	7,372 (36.6)
Chronic liver disease	42,085 (16.5)	413,957 (7.2)	2,426 (15.4)	1,497 (7.4)
Chronic kidney disease	56,424 (22.1)	219,492 (3.8)	3,771 (24.0)	1,168 (5.8)
Chronic gastrointestinal disease	124,625 (48.8)	2,138,827 (37.3)	7,267 (46.2)	6,630 (32.9)
Neurological disorders	104,910 (41.1)	813,170 (14.2)	6,926 (44.0)	3,585 (17.8)
Diabetes	12,350 (4.8)	76,901 (1.3)	731 (4.7)	327 (1.6)
Immunocompromising conditions+	38,730 (15.2)	322,796 (5.6)	2,142 (13.6)	1,376 (6.8)
Obesity	52,058 (20.4)	745,134 (13.0)	3,225 (20.5)	2,572 (12.8)

Figure 3. Modeled Mean Incidence Rate of Norovirus AGE by Age Group and Underlying Medical Condition, Across Settings

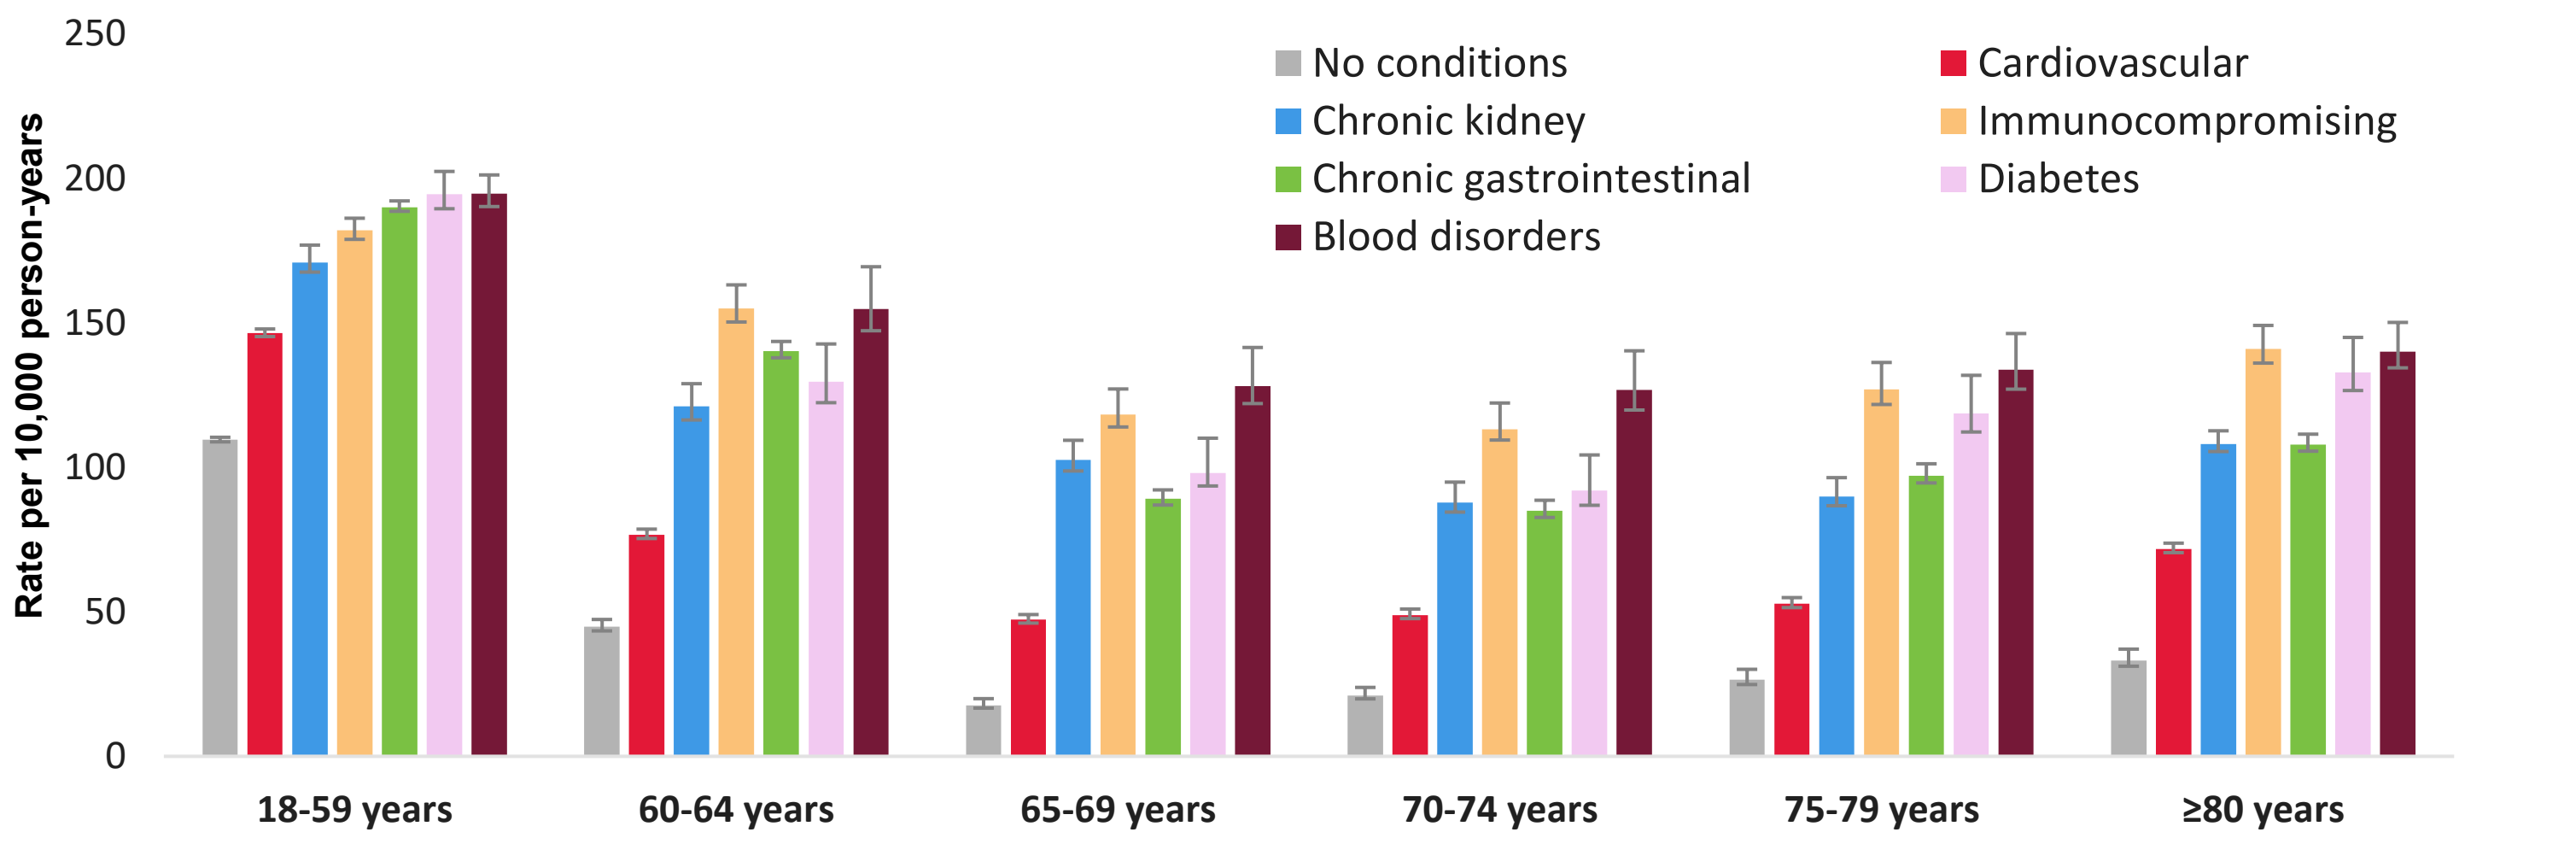
Rate per 10,000 person-years
250
200
150
100
50
0
No conditions
Cardiovascular
Chronic kidney
Immunocompromising
Chronic gastrointestinal
Diabetes
Blood disorders
18-59 years
60-64 years
65-69 years
70-74 years
75-79 years
≥80 years

- A total of 5.99 million all-cause AGE and 35,876 norovirus AGE episodes were identified from 2015–2024 (Table 1).
- The indirect model attributed 16% of cause-unspecified AGE to norovirus overall, resulting in approximately 27 additional cases for each identified cause-specified norovirus AGE episode.
- Incidence of inpatient admissions for AGE was highest among adults ≥80 years (Overall rate: 230 episodes/10,000 person-years [PY]) and incidence of outpatient AGE visits was highest among adults 18–59 years (Overall rate: 1,393/10,000 PY) (Figure 1).
- Norovirus AGE incidence followed similar patterns with highest inpatient incidence among adults ≥80 years (48/10,000 PY) and highest outpatient incidence among adults 18–59 years (226/10,000 PY) (Figure 2). Inpatient admissions occurred most frequently from November–March (63% of cases) particularly for adults ≥75 years. Outpatient trends were more heterogeneous across age groups, with modest increases in October–November, a December–January peak, and variable declines thereafter.
- Across all ages, adults with underlying conditions had substantially higher norovirus AGE incidence than those without comorbidities (Figure 3). The highest rates were observed among those with blood disorders, diabetes, and immunocompromising conditions, as well as among younger adults across all conditions.

CONCLUSIONS

- High outpatient incidence among younger adults demonstrates that the norovirus AGE and all-cause AGE burden extends beyond older populations, while hospitalization rates were highest among adults ≥80 years, highlighting age-related differences in severity. This pattern was more pronounced among norovirus AGE encounters, reflecting the substantial under-ascertainment inherent in claims-based cause-specified diagnoses alone. Age-related temporal shifts were also observed.
- Norovirus AGE incidence was consistently higher among adults with underlying medical conditions, specifically blood disorders (e.g., sickle cell disorders, nutritional anemias, coagulation and platelet disorders), diabetes, and immunocompromising conditions.
- These findings are consistent with prior estimates demonstrating substantial medically attended norovirus AGE burden across healthcare settings, and highest hospitalization rates among older adults.[3,4] The indirect modeling approach further suggests that reliance on cause-specified codes for norovirus alone greatly underestimates the true incidence of norovirus AGE, which should be an important consideration when estimating burden, especially among high-risk sub-populations.
- This study highlights the substantial clinical and healthcare burden of norovirus AGE in adults and reinforces the need for targeted preventive strategies, particularly for older adults and individuals with underlying medical conditions.

References

1. Burke RM, Mattison CP, Pindyck T, Dahl RM, Rudd J, Bi D, Curns AT, Parashar U, Hall AJ. Burden of Norovirus in the United States, as Estimated Based on Administrative Data: Updates for Medically Attended Illness and Mortality, 2001-2015. Clin Infect Dis. 2021 Jul 1;73(1):e1-e8. doi: 10.1093/cid/ciaa438. PMID: 32291450; PMCID: PMC8112883.
2. Burke, R.M. and A.J. Hall, Global Burden of Norovirus, in Norovirus. 2019. p. 1-29.
3. Verstraeten T, Jiang B, Weil JG, Lin JH. Modelling Estimates of Norovirus Disease in Patients with Chronic Medical Conditions. PLoS One. 2016 Jul 20;11(7):e0158822. doi: 10.1371/journal.pone.0158822. PMID: 27438335; PMCID: PMC4954678.
4. Kowalzik F, Binder H, Zöller D, Riera-Montes M, Clemens R, Verstraeten T, Zepp F. Norovirus Gastroenteritis among Hospitalized Patients, Germany, 2007-2012. Emerg Infect Dis. 2018 Nov;24(11):2021-2028. doi: 10.3201/eid2411.170820. PMID: 30334712; PMCID: PMC6199990.

Acknowledgments

This study was funded by Moderna, Inc.

Disclosures

This study was funded by Moderna, Inc. CK, BP, JR, SS, CV, SI, and KC are employees of Moderna, Inc. and hold stock/stock options in the company. RN and VV are employees of InGef GmbH, which acted as subcontractor and received funding from WIG2 for execution of the analysis.

moderna

ESCMID Global; April 17–21, 2026; Munich, Germany